Long-Term Efficacy and Safety of Abacavir/Lamivudine (ABC/3TC) with Fosamprenavir + Ritonavir Versus Lopinavir/Ritonavir (LPV/r) over 144 Weeks

Poster Number H1243

V. ESTRADA[1], K. LOGUE[2], P. DOMINGO[3], K. SCHEWE[4], L. YAU[5], C. VAVRO [5], C. PHARO[6], M. L. LIM[7]

[1]Hospital Clínico San Carlos, Madrid, Spain, [2] St. Clair Medical Associates, Toronto, Canada [3] Hospital de la Santa Creu i Sant Pau, Barcelona, Spain,[4] IPM-Studycenter, Hamburg, Germany, [5]GSK, RTP, NC, USA, [6]GSK, Brentford and [7]GSK Stockley Park, UK

Abstract

Background: The KLEAN study extension assessed the long term efficacy and safety of fosamprenavir-ritonavir (FPV/r) and lopinavir-ritonavir (LPV/r), both given with abacavir/lamivudine (ABC/3TC) fixed dose combination (FDC), over 144 wks.

Method: KLEAN is an open-label, non-inferiority study which randomized antiretroviral naïve patients to receive FPV/r twice daily (BID) or LPV/r BID with ABC/3TC FDC once daily (QD). Patients with a viral load <400 copies/mL at Wk 48 were eligible to participate in the KLEAN study extension (up to 144 wks).

Results: 196 patients participated in the KLEAN study extension (48-144 wks) and continued with their previously randomised therapy. The proportion of TLOVR responders (HIV-1 RNA <50 copies/mL) at Wk 144 was 73% in the FPV/r arm and 60% in the LPV/r arm. The proportion of responders (<50 copies/mL) was the same irrespective of baseline HIV-1 RNA (>100,000 or ≤100,000 copies/mL). The Wk 144 median [interquartile range] change from baseline CD4+ cell count was 300 [236–433] cells per μL and 335 [225–444] cells per μL in the FPV/r and LPV/r arms, respectively. Diarrhoea was the most frequently reported adverse event (AE). A small proportion of patients (13% in the FPV/r arm and 9% in the LPV/r arm) discontinued study medication due to AEs. Three patients (FPV/r: 1; LPV/r: 2) had virological failure between Wk 48 and Wk 144.

Conclusion: FPV/r and LPV/r, when used in combination with the ABC/3TC fixed dose combination backbone, show durable viral load suppression over 144 weeks, including in both high and low baseline viral load strata.

Introduction

- The KLEAN study was conducted in antiretroviral-naïve patients and designed to demonstrate non-inferiority of the antiviral efficacy, safety and tolerability of twice daily fosamprenavir-ritonavir (FPV/r) and lopinavir-ritonavir (LPV/r) both administered with abacavir/lamivudine (ABC/3TC) fixed dose combination (FDC) [1].
- At 48 wks, the study confirmed that FPV/r is non-inferior to LPV/r in terms of antiretroviral efficacy, safety, and tolerability. US and European agencies subsequently recommend the use of FPV/r as a preferred protease inhibitor component of initial antiretroviral therapy regimens for treatment of antiretroviral-naïve patients.
- Despite FPV/r having demonstrated non-inferiority to LPV/r for treatment up to 1 year, longer periods of safety and efficacy confirmed with LPV/r have not yet been established for FPV/r.
- The purpose of the KLEAN study extension was to assess the long term efficacy and safety of FPV/r and LPV/r, both administered with ABC/3TC FDC, over 144 wks.

Methods

- The KLEAN study was a Phase IIIb, randomized, open-label, international, multi-centre non-inferiority trial. Patients were randomized to one of two oral antiretroviral regimens (A or B) in a 1:1 ratio. Randomization was stratified at entry according to screening plasma HIV-1 RNA levels <100,000 and ≥100,000 copies/mL.
 - Treatment Arm A: one FPV (GlaxoSmithKline, NC, USA) 700 mg tablet BID plus one ritonavir (Abbott Laboratories, Illinois, USA) 100 mg capsule BID plus one ABC/3TC 600 mg/300 mg (GlaxoSmithKline, NC, USA) FDC tablet QD.
 - Treatment Arm B: LPV/r 400 mg/100 mg (Abbott Laboratories, IL, USA) given as three LPV/r 133.3/33.3 mg capsules BID plus one ABC/3TC 600 mg/300 mg FDC tablet QD.
- At 48 wks, patients that had maintained viral suppression to <400copies/mL, and who were willing to participate in the KLEAN study extension (48-144wks) underwent a KLEAN study extension baseline evaluation. Those who fulfilled the study eligibility criteria continued with their existing study medication into the extension phase of the study.
- Patients were evaluated for safety, tolerability and efficacy (HIV-1 RNA, CD4+ and CD8+ analyses) at the baseline extension phase visit, and at wks 60, 72, 84, 96, 108, 120, 132 and 144 (or at early withdrawal).
- The primary population for efficacy analyses at Wk 144 was the Intent-to-Treat-Extension (ITT-Ext) population and the primary population for safety analyses was the Safety-Extension population (where patients were analyzed according to the treatment actually received).
- Efficacy endpoints consisted of the proportions of patients with plasma HIV-1 RNA <400 copies/mL and <50 copies/mL at Wk 144, and the absolute and change from baseline values in plasma HIV-1 RNA and CD4+ cell counts.
- Due to limitations in the design of the KLEAN study extension (48 to 144 weeks), including reduced sample size and selection bias of patients who participated; it was not powered to test for non-inferiority between the two treatments.

Results

- 196 patients took part in the KLEAN extension. Demographic characteristics were similar between the treatment arms (Table 1). Median [range] exposure to FPV/r was 144 [60–153] wks and to LPV/r was 143 [32–151] wks. Thirty-four (17%) patients withdrew from the study extension prematurely, 2 as confirmed virological failure, 2 due to insufficient viral load response, 8 due to adverse events and 22 due to other reasons.
- The median baseline HIV-1 RNA value for all patients who took part in the KLEAN study extension (48–144 wks) was high (5.0 $\log_{10}$ [range: 2.95–6.77] copies/mL) and the median value for CD4+ cell count was low (203 [range: 19–837] cells per μL). CD4+ cell counts for the two treatment groups at baseline were generally similar, the proportion of patients in the ITT-Ext population with baseline HIV-1 RNA ≥100,000 copies/mL was higher in the FPV/r arm than the LPV/r arm (57% and 47%, respectively) (Table 1).
- The proportion of patients with HIV-1 RNA <400 copies/mL declined slightly over time after Wk 48 of the study. For the proportion of patients with HIV-1 RNA <50 copies/mL, both treatment arms showed a decline over time from Wk 48 of the study, the decline being numerically greater in the LPV/r arm at 72 wks onwards (Figure 1). Protocol-defined virological failure during the extension was similar between treatment groups (FPV/r <1% and LPV/r 2%, respectively).

Figure 1. Proportions of patients (ITT-Ext) with HIV-1 RNA <400 copies/mL and <50 copies/mL (TLOVR analysis)

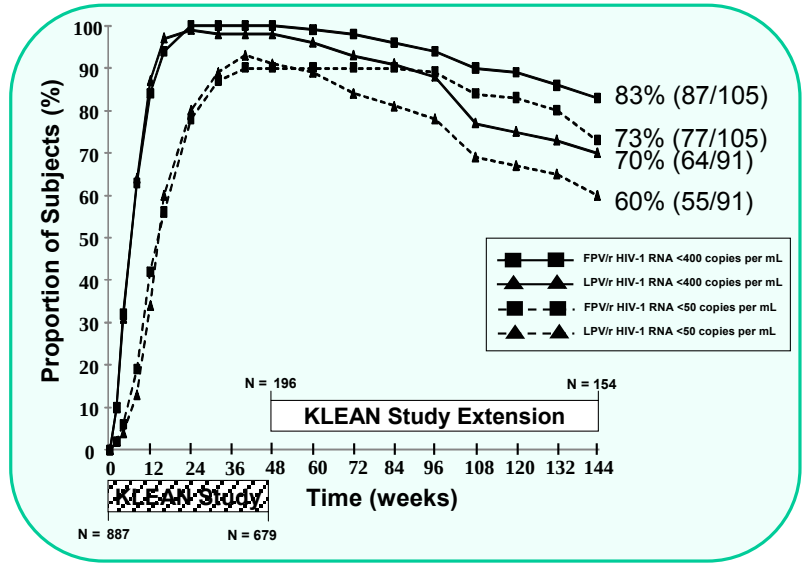
Proportion of Subjects (%)
Time (weeks)
83% (87/105)
73% (77/105)
70% (64/91)
60% (55/91)
FPV/r HIV-1 RNA <400 copies per mL
LPV/r HIV-1 RNA <400 copies per mL
FPV/r HIV-1 RNA <50 copies per mL
LPV/r HIV-1 RNA <50 copies per mL
N = 196
N = 154
KLEAN Study Extension
KLEAN Study
N = 887
N = 679

Table 1. Baseline characteristics, ITT-Extension population

	FPV/r (N=105)	LPV/r (N=91)
Sex; n female (%)	26 (25%)	18 (20%)
Age in years; median [min-max]	38.0 [20–78]	38.0 (21–74]
HIV-1 RNA $\log_{10}$ copies per mL; median (IQR)	5.12 (4.73–5.62)	4.95 (4.66–5.41)
HIV-1 RNA ≥100,000 copies per mL; n (%)	60 (57%)	43 (47%)
CD4 cells per μL; median (IQR)	197 (111–280)	213 (103–288)
CDC classification C; n (%)	12 (11%)	11 (12%)
Hepatitis B positive; n (%) *	3 (3%)	2 (2%)
Hepatitis C positive; n (%) *	9 (9%)	4 (4%)
HIV risk factors, n (%)		
Homosexual contact	54 (51%)	46 (51%)
Heterosexual contact	43 (41%)	41 (45%)
Injectable drug use	5 (5%)	0
Transfusion	1 (<1%)	1 (1%)
Other	4 (4%)	1 (1%)

*There were no patients who had both hepatitis B and C.

FPV/r = fosamprenavir/ritonavir; LPV/r = lopinavir/ritonavir; IQR = interquartile range.

Figure 2. Proportions of patients with HIV-1 RNA <400 (a) and <50 (b) copies/mL (TLOVR analysis) at Wk 144 by baseline viral load

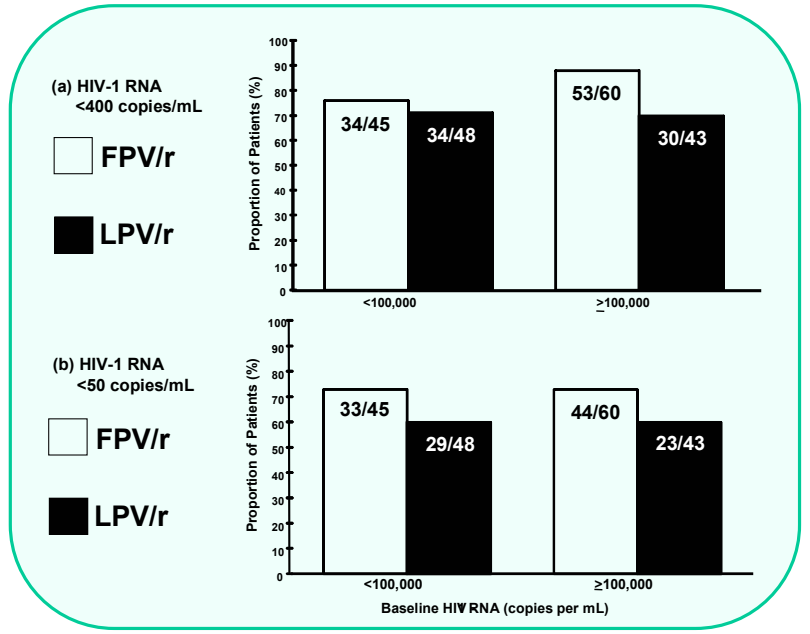
(a) HIV-1 RNA <400 copies/mL
FPV/r
LPV/r
Proportion of Patients (%)
34/45
34/48
53/60
30/43
<100,000
≥100,000
(b) HIV-1 RNA <50 copies/mL
FPV/r
LPV/r
33/45
29/48
44/60
23/43
Baseline HIV RNA (copies per mL)

- High viral load at baseline (≥100,000 copies/mL) did not adversely affect viral response rates in either treatment arm (Figure 2a [<400 copies/mL] and Figure 2b [<50 copies/mL]).
- Median CD4+ cell count at Wk 144 was 546 cells/μL in the FPV/r arm and 550 cells/μL in the LPV/r arm. Median (IQ range) change from baseline CD4+ cell count at Wk 144 was +300 (236 to 433) cells/μL in the FPV/r arm and +335 (225 to 444) cells/μL in the LPV/r arm. Progression of HIV disease, as defined by CDC Category A to Category C, occurred in one patient in the LPV/r treatment arm. Progression from CDC Category C to New CDC Category C occurred in one patient in the FPV/r arm.
- AEs were reported at similar overall numbers in both treatment arms (Table 3). 186 patients reported at least one AE irrespective of causality, 101 (97%) in the FPV/r arm and 85 (92%) in the LPV/r arm. Most AEs were rated as either grade 1 or grade 2 in intensity. Eight patients (FPV/r 5 (5%); LPV/r 3 (3%)) experienced AEs that led to premature discontinuation from the study. Changes from baseline in fasting total cholesterol, LDL-c, HDL-c, triglycerides, and insulin were similar across treatment groups and analysis of renal and hepatic function did not reveal any changes of clinical concern from baseline through Wk 144.

Table 3. Number (%) of drug-related adverse events (grades 2–4) and laboratory abnormalities (grades 3–4) in ≥2% of patients through 144 weeks, Safety-Extension population

	FPV/r (N=104)	LPV/r (N=92)
Any Event	**44 (42%)**	**25 (27%)**
Clinical adverse event		
Diarrhoea*	16 (15%)	11 (12%)
Drug hypersensitivity	8 (8%)	5 (5%)
Nausea	6 (6%)	2 (2%)
Hypercholesterolemia	5 (5%)	1 (1%)
Abdominal pain upper	3 (3%)	2 (2%)
Blood cholesterol increased	4 (4%)	1 (1%)
Hypertriglyceridemia	2 (2%)	2 (2%)
Rash	4 (4%)	0
Abdominal distension	3 (3%)	0
Abdominal pain	3 (3%)	0
Blood triglycerides increased	3 (3%)	0
Vomiting	0	3 (3%)
Laboratory abnormalities†		
Fasting triglycerides	7/81 (9%)	6/77 (8%)
Creatine kinase	8/104 (8%)	7/92 (8%)
Fasting cholesterol	5/81 (6%)	4/77 (5%)
Alanine Amino Transferase	4/104 (4%)	1/92 (1%)
Aspartate aminotransferase	2/104 (2%)	1/92 (1%)
Glucose	1/104 (<1%)	2 (2%)
Potassium	1/104 (<1%)	2 (2%)

* When only counting adverse events with onset after week 48, diarrhoea was the only drug-related Grade 2–4 event that occurred in ≥2% of patients: 3% in the FPV/r arm and 4% in the LPV/r arm. †Denominators show number of patients tested. PV/r = fosamprenavir/ritonavir; LPV/r = lopinavir/ritonavir.

Discussion

- The KLEAN study 48 wk data established that FPV/r combination treatment is non-inferior to LPV/r over 48 wks when used with the same once-daily NRTI backbone of ABC/3TC FDC [1]. It also showed a potent antiretroviral effect in those with both high and low baseline viral loads as well as in those with low baseline CD4+ cell counts (<50 cells/μL) when used in combination with the ABC/3TC fixed dose combination.
- This study reports a long-term extension of up to 3 yrs to a well-powered comparison of two ritonavir-enhanced protease inhibitors with ABC/3TC in HIV-1 infected treatment-naïve patients. It supports potency and durability of viral suppression with FPV/r similar to that seen with LPV/r over 3 years, in both high and low baseline viral loads, when used in combination with the ABC/3TC fixed dose combination.
- Absolute and change from baseline in HIV-1 RNA and CD4+ cell counts were similar in the FPV/r and LPV/r treatment arms with the proportion of patients with HIV-1 RNA <50 copies/mL and <400 copies/mL in the FPV/r arm being numerically higher than the proportion with HIV-1 RNA <50 copies/mL and <400 copies/mL in the LPV/r arm, respectively.
- Both the FPV/r and LPV/r regimens were generally safe and well tolerated when used in combination with ABC/3TC FDC over 144 wks. The proportion of patients who discontinued study due to AEs, who experienced changes of clinical concern in renal or hepatic function or increases in fasting serum lipids were similar across treatment arms.
- Drug-associated resistance was rare and no additional major protease-associated resistance was detected in the extension phase to 144 wks.

Conclusions

- The KLEAN study extension (48 to 144 weeks) supports durable viral suppression with FPV/r and LPV/r when used in combination with ABC/3TC over 144 weeks.
- Viral suppression was comparable between those patients who started with high baseline viral loads of greater than 100,000 copies/mL and lower viral loads with both treatment arms.
- Only three patients experienced virological failure during the study extension (from Week 48 to Week 144), and no protease associated drug resistance was detected during this time when FPV/r and LPV/r were used in combination with the ABC/3TC.
- Both regimens were safe and generally well tolerated.

Acknowledgements

The authors would like to thank all participating study subjects, site personnel and GSK study teams.

References

1. Eron J Jr, et al. KLEAN study team. The KLEAN study of fosamprenavir-ritonavir versus lopinavir-ritonavir, each in combination with abacavir-lamivudine, for initial treatment of HIV infection over 48 weeks: a randomised non-inferiority trial. *Lancet*. 2006;368:476-82.

48th Annual Interscience Conference on Antimicrobial Agents and Chemotherapy Meeting, 25–28 October 2008, Washington, DC, USA